A Randomized Clinical Trial to Evaluate the Immunogenicity and Safety of ROTAVAC® in comparison with a licensed rotavirus vaccine in healthy infants

Ella R, Prasad S, Krishna Mohan V
Bharat Biotech International Limited, Hyderabad, India

Background

- The World Health Organization (WHO) recommends that rotavirus (RV) vaccines should be included in national immunization programs.
- ROTAVAC® (nHRV), derived naturally from the human 116E neonatal strain, was licensed in India in 2015 based on promising results of a phase 3, safety and efficacy trial.
- As a pre-requisite for WHO prequalification, we compared the immunogenicity and safety of ROTAVAC® to those of a WHO-prequalified, Rotarix®.

Methods

- We conducted a multicentre, open-labeled, randomized phase 4 clinical trial where 464 infants, 6 to 8 weeks of age were equally randomized to receive as licensed, the complete regimen of ROTAVAC® (3 doses; Group I) or Rotarix® (2 doses; Group II).
- Antibody responses (serum anti-RV Immunoglobulin A [IgA]) were measured by enzyme-linked immunosorbent assay (ELISA). The primary analysis was an assessment of non-inferiority of ROTAVAC® to Rotarix® for geometric mean titer (GMT) for infants who received the complete regimen of either vaccine.

Results:

Conclusions:

Limitations:

Clinical Trials Registration: (CTRI Number: CTRI/2015/12/006428).